ESMO 2018: Poster #437P

PT-112: A Well-Tolerated Novel Immunogenic Cell Death (ICD) Inducer with Activity in Advanced Solid Tumors

Daniel D. Karp[1], D. Ross Camidge[2], Jeffrey Infante[3], Tyler D. Ames[4], Jose Jimeno[4], and Alan H. Bryce[5]

[1]The University of Texas MD Anderson Cancer Center, Houston, TX; [2]University of Colorado Cancer Center, Aurora, CO; [3]Sarah Cannon Research Institute, Nashville, TN; [4]Phosplatin Therapeutics, New York, NY; [5]Mayo Clinic, Scottsdale, AZ

Study sponsored by Phosplatin Therapeutics

Chemical Structure of PT-112

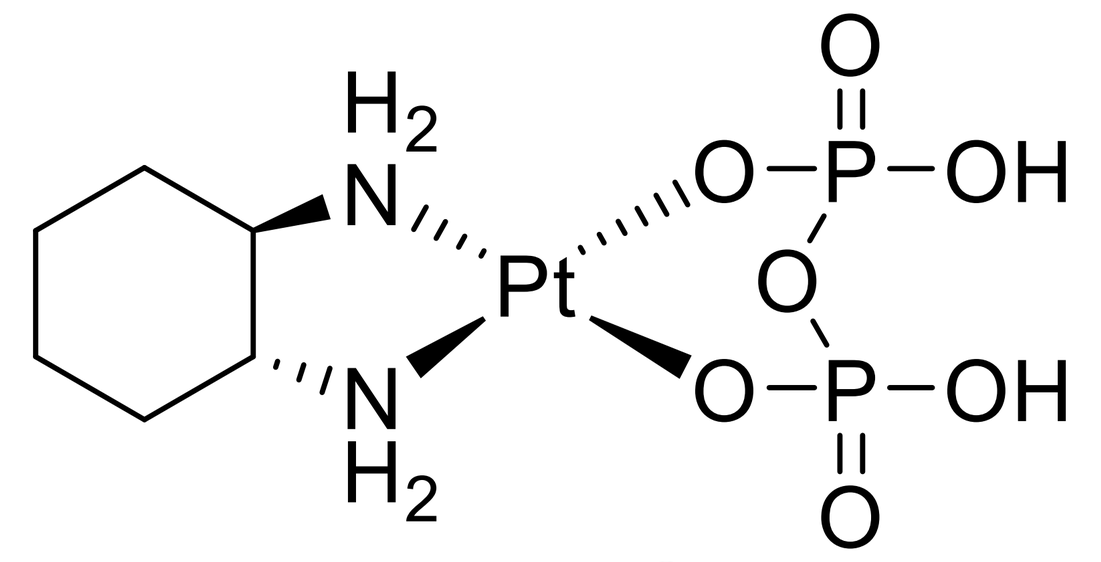

H2
N
Pt
O−P−OH
O
O−P−OH
N
H2
O
O

PT-112

Background / Rationale

- PT-112 is a novel chemical entity with numerous interesting and beneficial properties observed in preclinical development
- Minimal DNA binding and a lack of sensitivity to DNA damage repair pathways *in vitro* indicate DNA is not the primary mechanistic target of PT-112[1]
- Effects on apoptosis and cell cycle proteins observed *in vitro* include p53, MDM2, p16 and downstream CDKs
- Biodistribution profile in mice demonstrates delivery of active concentrations to tissues that are important sites of disease in advanced cancer patients, notably lung, liver, and bone tissues[2]
- Robust induction of immunogenic cell death (ICD) has been observed *in vitro* and *in vivo:*
 - Release of damage-associated molecular patterns (DAMPs), indicative of ICD induction
 - Large increases in T cell populations in tumor tissue in murine models
 - Robust evidence of synergy in murine allograft models in combination with checkpoint inhibitors
 - 100% tumor-free survival in murine ICD vaccination models, the gold standard for ICD validation
- These data suggest PT-112 may potentially be a best-in-class ICD inducing agent[3]
- Exceptional safety profile differs notably from conventional chemotherapeutic compounds

Methods / Study Design

A 3+3 study design was used to determine safety, tolerability, and the recommended phase II dose (RP2D) of PT-112 for patients with advanced solid tumors. Pharmacokinetics (PK) and anti-tumor effects were also assessed. PT-112 was given IV for 1 hr on days 1, 8, and 15 of a 28 day cycle, starting at a dose of 12 mg/m^2. Supplemental enrollment at dose levels deemed safe was allowed for the purpose of gathering additional data.

Results

- 11 dose levels assessed, ranging from 12-420 mg/m^2, all deemed safe; MTD not reached
- RP2D declared at 360 mg/m^2 on the basis of anti-tumor activity, safety, and dose-linear PK profile
- Mild fatigue most common side effect; limited myelosuppression and neurotoxicity
- Tumor control, biomarker, and metabolic responses observed at doses ≥125 mg/m^2
- Durable partial responses (PRs) achieved in thoracic tumors starting at 250 mg/m^2, including in patients resistant or refractory to immune checkpoint inhibitors

Demographics and Dose Levels

Table 1: PT-112-101 Patient Demographics Summary

Characteristic	Number	Percentage
Total Number of Pts	66	100
Age (years)		
Median	66	
Range	25-86	
Gender		
Male	38	58
Female	28	42
Ethnicity		
Caucasian	54	82
African American	9	14
Asian	2	3
Other	1	1
ECOG PS		
0	19	29
1	47	71
Tumor Types		
Appendiceal	2	3
Basal Cell	1	2
Bladder	1	2
Breast	2	3
Cervical / Uterine	3	5
Colorectal	6	9
Endometrial	3	5
Esophageal	1	2
Gastric	1	2
Head and Neck	13	20
Lung	6	9
Melanoma	2	3
Mesothelioma	1	2
Neuroendocrine	1	2
Osteosarcoma	2	3
Ovarian	5	8
Pancreatic	2	3
Prostate	10	16
Skin	1	2
Thymoma	2	3
Thyroid	1	2
Sites of Disease		
Lung	28	42
Liver	21	32
Lymph Node	18	27
Bone	14	21
Other	36	55
Number of Sites		
Single	29	44
Multiple	37	56
Median Prior Lines of Systemic Therapy	4	

Tumor types in which objective evidence of biological activity has been observed (highlighted: Basal Cell, Gastric, Head and Neck, Lung, Ovarian, Pancreatic, Prostate, Thymoma)

Table 2: Patient Exposure and Disposition

Highest Dose Received (mg/m^2)	12-96	125	150	200	250	300	360	420
Treated, n	12	3	10	5	16	10	7	3
Median number of infusions, n (range)	6 (1-60)	13.5 (3-21)	4.5 (3-21)	5 (2-9)	6 (1-23)	3 (1-10)	6 (3-14)	6 (3-12)
Continuing treatment, n	0	0	0	0	0	0	2	0
Partial responses, n	0	0	0	0	1	0	2	0
PFS @ 6 months, n	1	2	1	0	3	0	2	0
Best response of SD or better, n	4	1	2	2	5	3	4	2
Reasons for treatment discontinuation, n	12	3	10	5	16	10	5	3
Disease progression	9	3	6	4	11	4	4	2
Treatment related AEs	0	0	3	1	4	2	1	0
Unrelated AEs	2	0	0	0	0	2	0	0
Other	1	0	1	0	1	2	0	1

Safety Profile of PT-112

Table 3: Adverse Events in ≥5% of Patients at Dose levels ≥125 mg/m2

Patients (n=54)	Treatment-Related AEs			
AE Term	All Grades		Grade ≥ 3*	
Fatigue	17	(31%)	1	(2%)
Nausea	13	(24%)	0	--
Peripheral Neuropathy	12	(22%)	2	(4%)
Thrombocytopenia	12	(22%)	4	(7%)
Vomiting	9	(17%)	0	--
Anemia	8	(15%)	3	(6%)
Neutropenia	7	(13%)	6	(11%)
Anorexia	7	(13%)	0	--
Diarrhea	6	(11%)	0	--
Constipation	6	(11%)	0	--
Acute Peripheral Neuropathy	4	(7%)	0	--
Hypophosphatemia	3	(6%)	2	(4%)
Dysgeusia	3	(6%)	0	--
Infusion Site Pain	3	(6%)	0	--

** No Grade 4 or 5 drug-related AEs were experienced*

Table 4: DLT Patient Characteristics

Parameter	Patient # 2-05-009	3-05-005	2-09-012	3-10-017
Dose Level (mg/kg)	150	150	250	300
Gender/Age	Female/75	Female/74	Female/34	Male/56
Tumor Type	Ovarian	Colon	Cervical	Head/Neck
DLT	Gr 3 Pancytopenia	Gr 3 Hypersensitivity Reaction	Gr 2 Renal Toxicity	Gr 3 Rash
No. of Prior Lines of Tx	6	4	1	3
Cis/Carbo/Oxali/ Taxane/Vinca	Carbo(x4)/ Taxane(x2)	Oxali	Cis/Taxane	Carbo
Radiotherapy	No	No	No	No
Pelvic Disease	Yes	No	Yes	No
Bone Marrow Function at Entry	Gr 2 Anemia; Gr 1 Neutropenia	Normal	Gr 2 Anemia	Normal
Renal Function at Entry	Normal	Normal	Elevated Creatinine	Low Creatinine
Liver Mets	No	Yes	No	No
Bone Mets	No	No	No	No
Pelvic Mets (Bulky)	No	No	No	No
Time of Event	7 days post C1D22	C1D15	9 days post C1D22	3 days post C1D8
Time to Recovery	14 days	1 day	12 days	21 days
Full Recovery	Yes	Yes	Yes	To Grade 1

Heavily Pretreated Patients with PFS ≥6 Months (9)

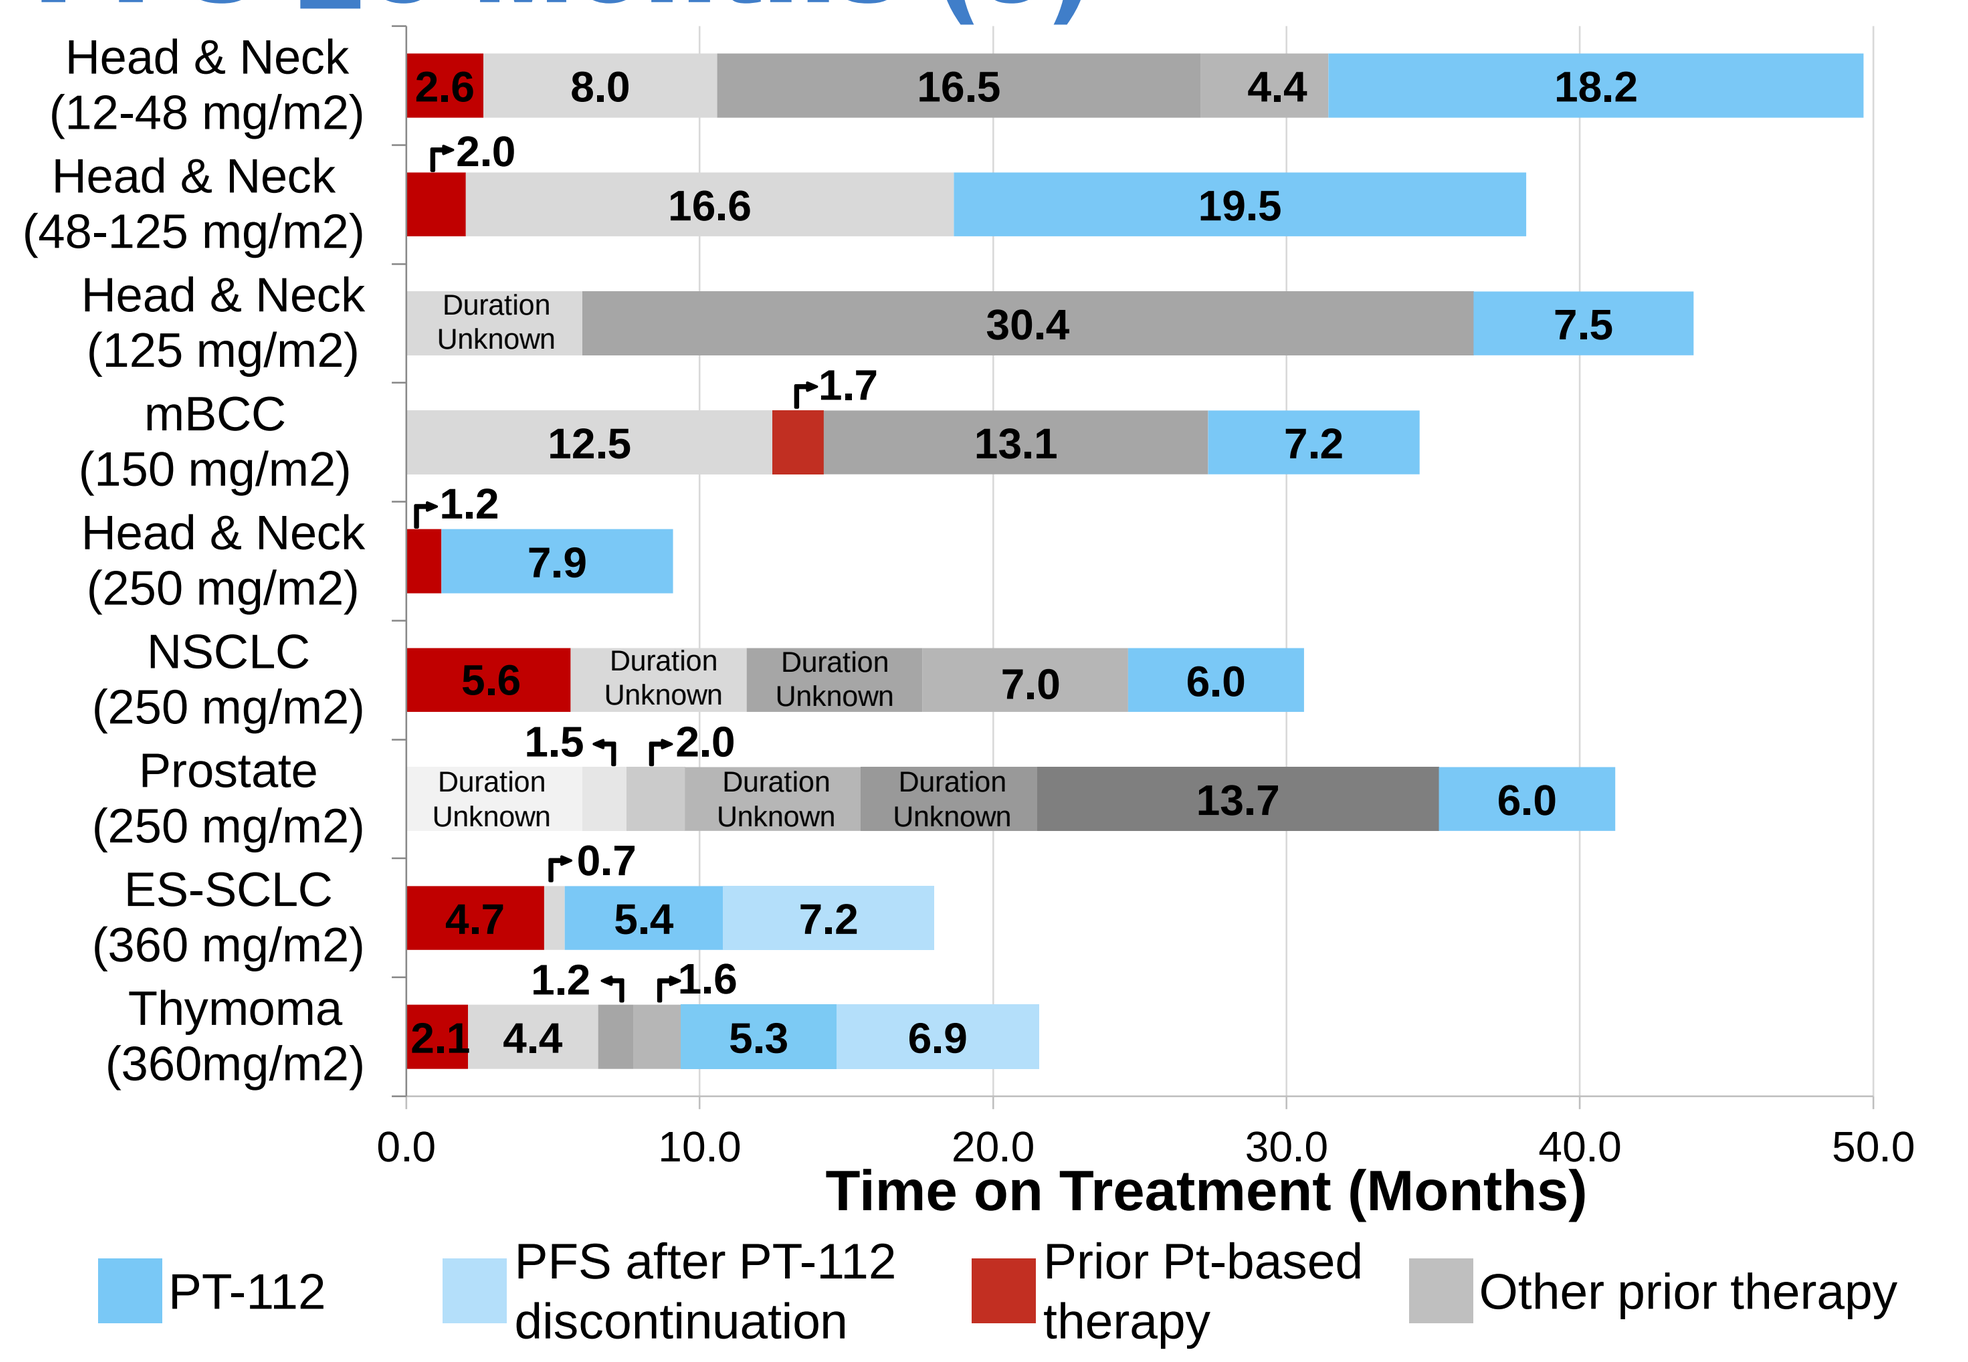

Head & Neck (12-48 mg/m2)
2.6
8.0
16.5
4.4
18.2
Head & Neck (48-125 mg/m2)
2.0
16.6
19.5
Head & Neck (125 mg/m2)
Duration Unknown
30.4
7.5
mBCC (150 mg/m2)
12.5
1.7
13.1
7.2
Head & Neck (250 mg/m2)
1.2
7.9
NSCLC (250 mg/m2)
5.6
Duration Unknown
Duration Unknown
7.0
6.0
Prostate (250 mg/m2)
Duration Unknown
1.5
2.0
Duration Unknown
Duration Unknown
13.7
6.0
ES-SCLC (360 mg/m2)
0.7
4.7
5.4
7.2
Thymoma (360mg/m2)
1.2
1.6
2.1
4.4
5.3
6.9
0.0
10.0
20.0
30.0
40.0
50.0
Time on Treatment (Months)
PT-112
PFS after PT-112 discontinuation
Prior Pt-based therapy
Other prior therapy

Figure 1: Treatment durations for PT-112 and previous treatments for patients with PFS ≥6 months on PT-112. 9 / 66 patients achieved PFS ≥6 months on the PT-112-101 study. There are two known cases where patients remained progression free for a long period of time after treatment discontinuation in the absence of PT-112 or any other treatment. All 9 patients had documented disease progression at study entry.

Durable PR (6 mos PFS) in NSCLC Patient, anti-PD-1 non-Responder

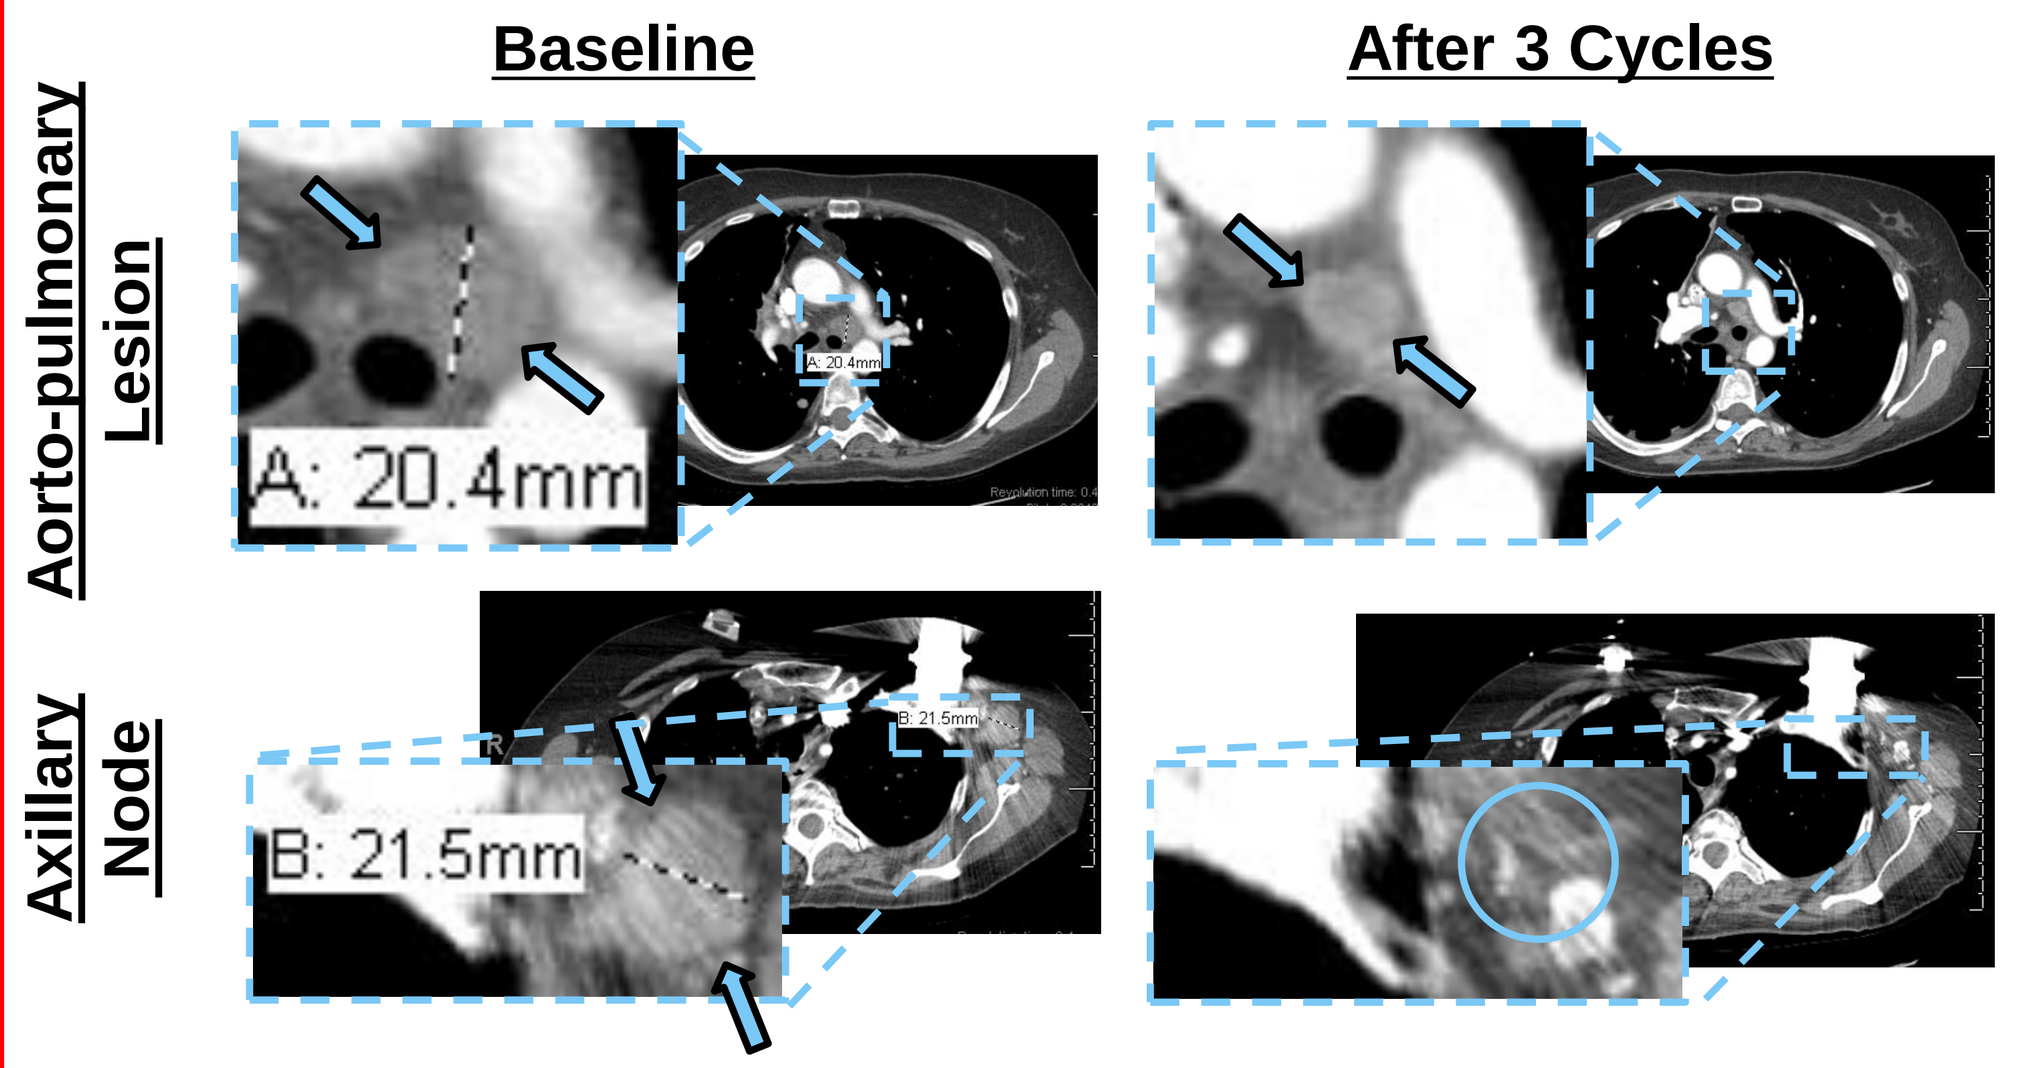

Baseline
After 3 Cycles
Aorto-pulmonary Lesion
Axillary Node
A: 20.4mm
B: 21.5mm

Figure 2: PR in a Post I-O NSCLC Patient. Patient received multiple lines of radiotherapy and four lines of systemic therapy, including 1st line Pt doublet, TKI, and anti-PD-1. Following response to 1st line treatment, best response to all subsequent treatments was stable disease (SD). A durable PR was achieved with PT-112 at a dose of 250 mg/m^2, first observed after 2 cycles, with tumor reductions reaching 39% as measured by RECIST 1.1. Complete metabolic response via PET was observed at primary lung as well as hepatic and bone metastatic sites (data not shown). This clinical case was previously presented[4].

Durable PR (>1 yr) in anti-CTLA4 + PD-1 Refractory ES-SCLC Pt

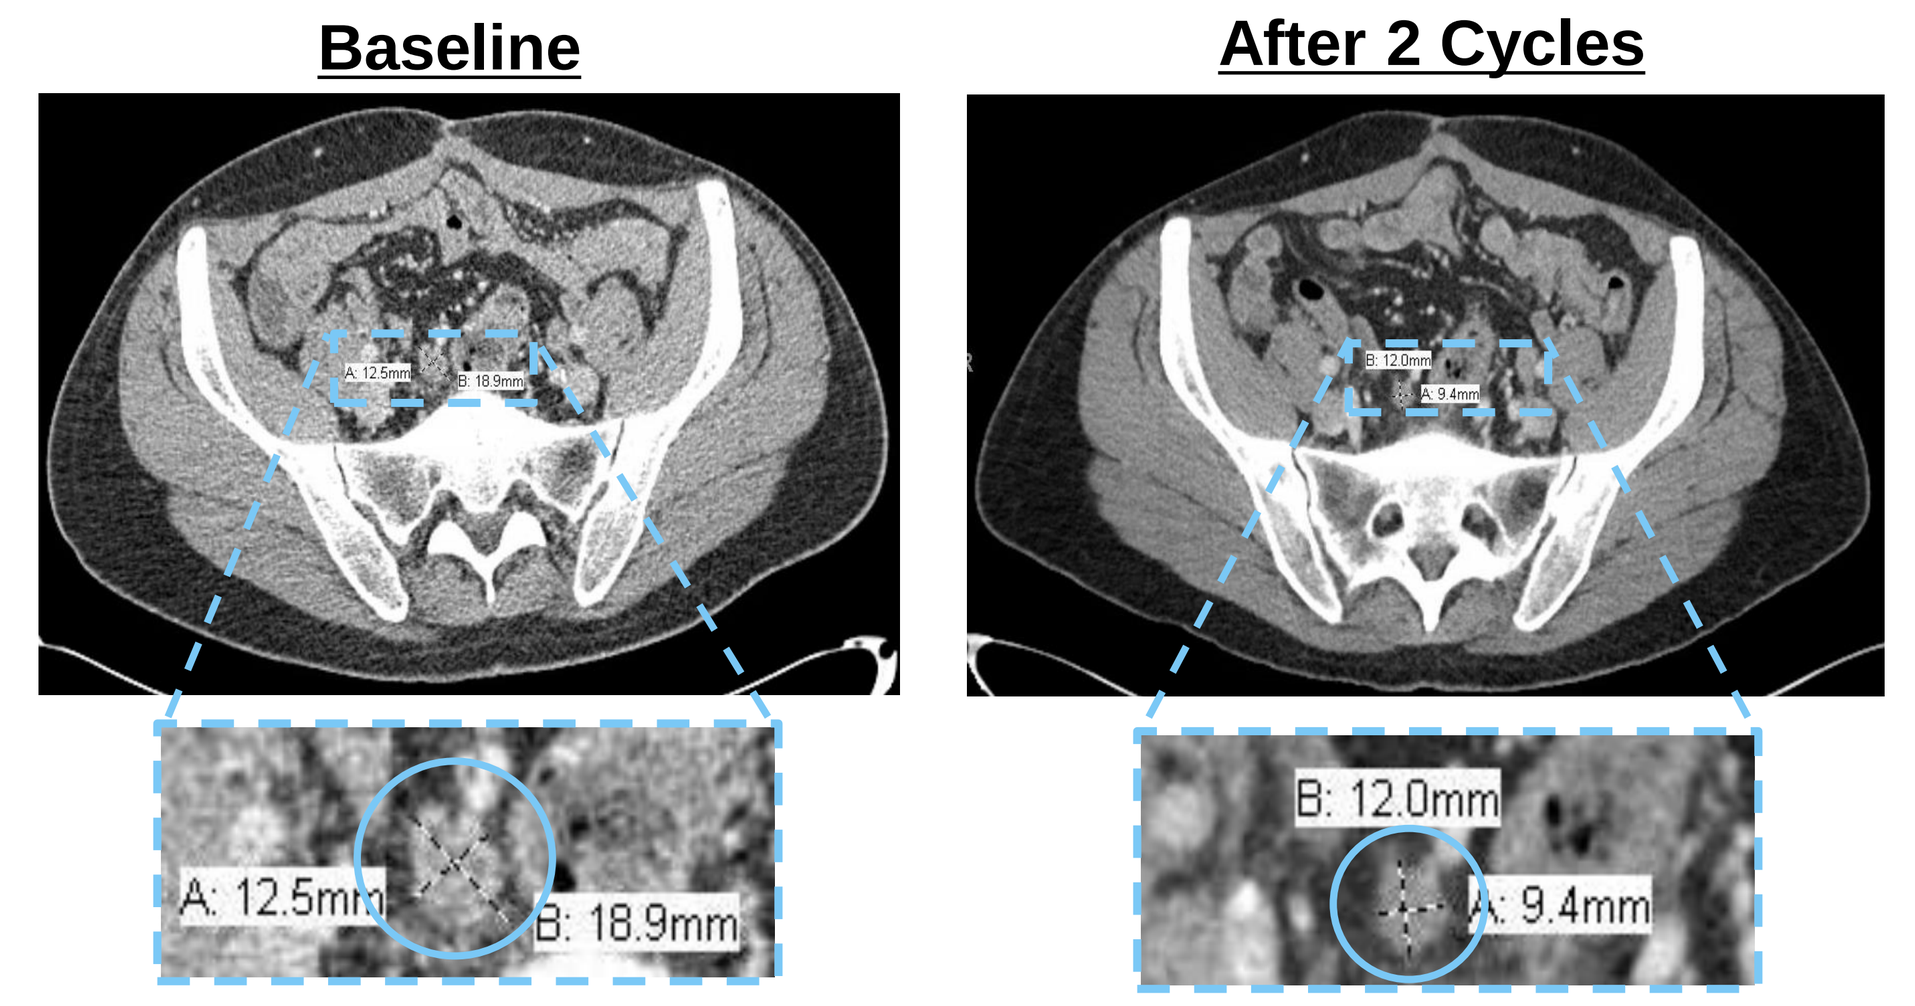

Baseline
After 2 Cycles
A: 12.5mm
B: 18.9mm
B: 12.0mm
A: 9.4mm

Figure 3: PR in a I-O Refractory ES-SCLC Patient. Patient experienced tumor shrinkage of 42% by RECIST 1.1, in addition to a 72% reduction in serum carcinoembryonic antigen levels, at the RP2D. Patient was previously treated with combination ipilimumab and nivolumab, with rapid progression noted after <1 month. Patient was treated on study for 5.4 months and remained progression free 7.2 months after treatment discontinuation, with continued tumor shrinkage at distal sites (not shown).

PET Response in Pancreatic Cancer Liver Metastasis

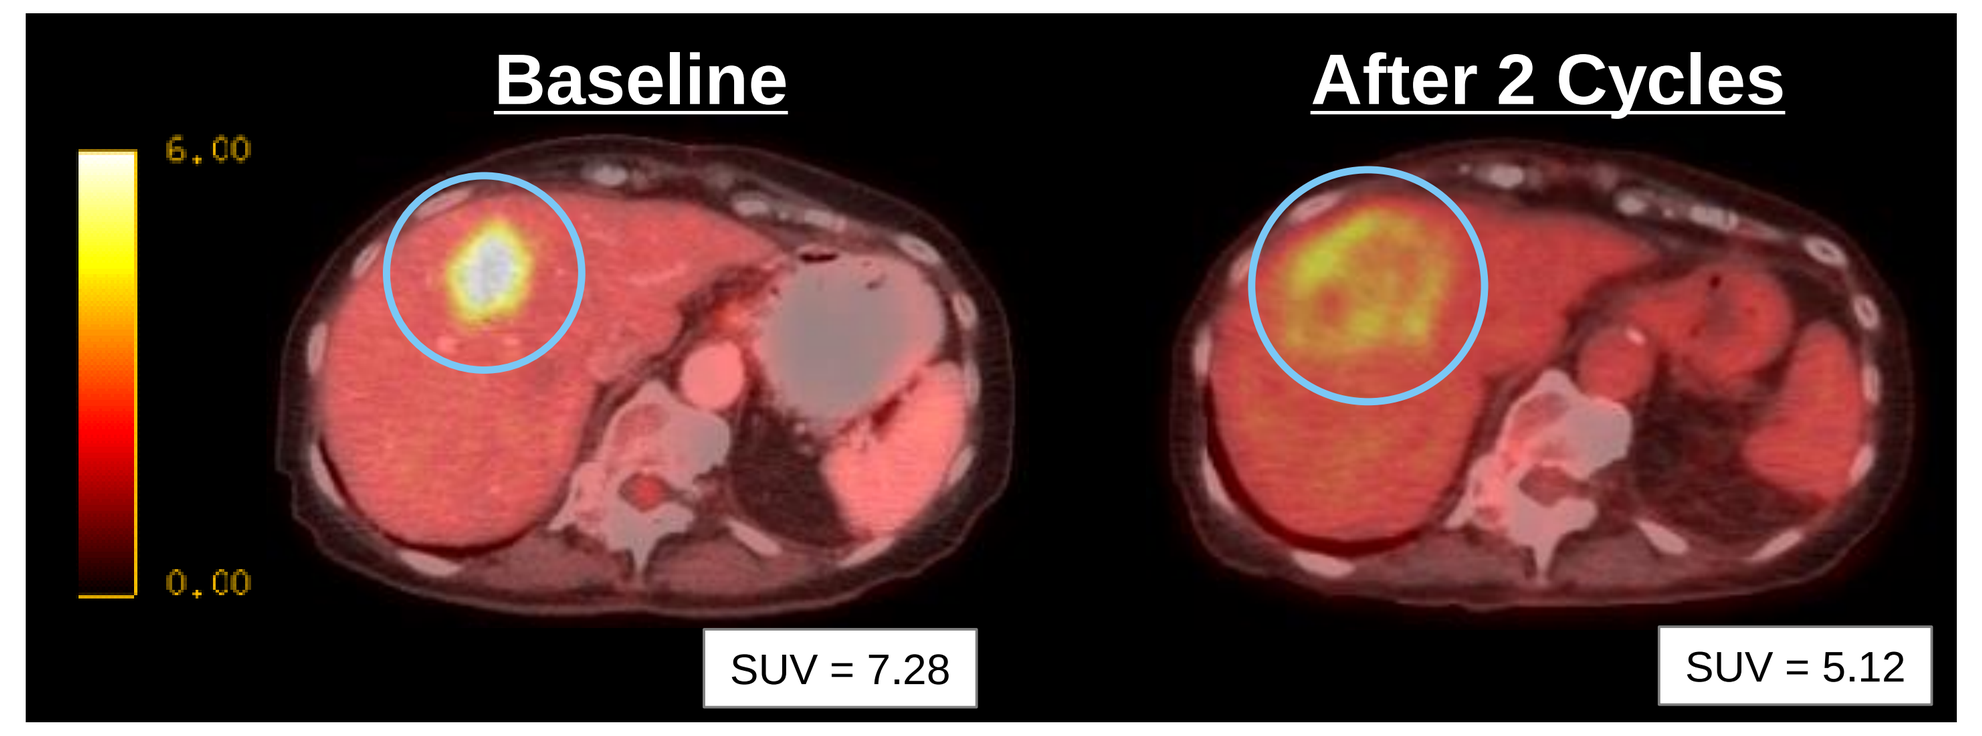

Baseline
After 2 Cycles
6.00
0.00
SUV = 7.28
SUV = 5.12

Figure 4: PET response in Pancreatic Cancer Patient. A metabolic response via PET was observed in a patient with pancreatic cancer after two cycles of treatment at a dose of 250 mg/m^2. The reduction in signal intensity, coupled with the hollowed out appearance, are indicative of tumor necrosis with immunological effects. This clinical case was previously presented[4].

Durable PR (>1 year PFS) in Advanced Thymoma Patient

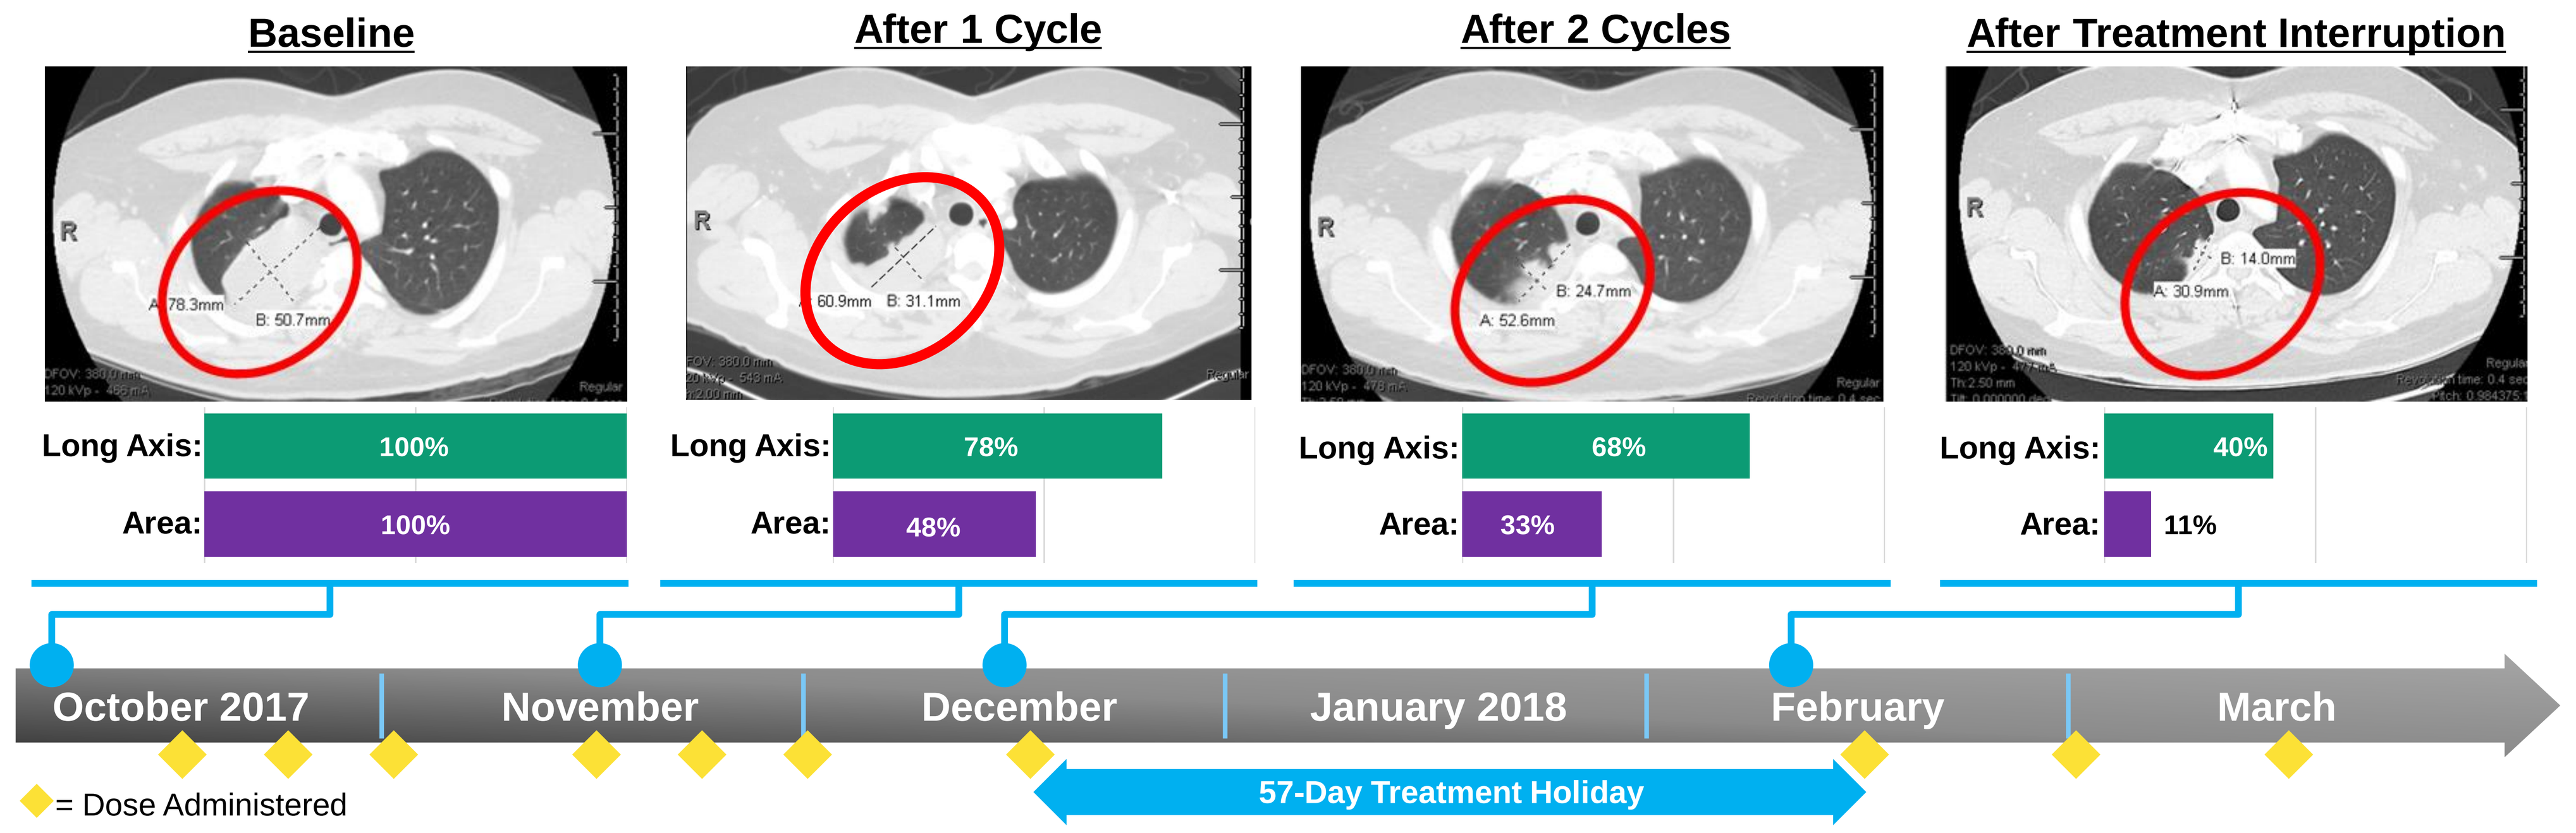

Baseline
After 1 Cycle
After 2 Cycles
After Treatment Interruption
Long Axis: 100%
Area: 100%
Long Axis: 78%
Area: 48%
Long Axis: 68%
Area: 33%
Long Axis: 40%
Area: 11%
October 2017
November
December
January 2018
February
March
= Dose Administered
57-Day Treatment Holiday

Figure 5: PR in an Advanced Thymoma Patient with 4 lines of prior treatment. Marked tumor shrinkage at pleural site of disease was observed at the RP2D over the first two cycles, representing a PR via modified RECIST SWOG criteria for pleural disease[5]. After a period of 8 weeks' treatment interruption, unrelated to disease or side effects, continued tumor shrinkage was observed. After a total of 5 cycles of treatment, at which point response was ongoing, therapy was suspended for observation. Patient remains progression free after 1 year. Disease stability is ongoing, with no therapy since March 2018. An additional thymoma patient experienced tumor necrosis and limited tumor shrinkage prior to treatment discontinuation (data not shown).

Examples of PSA Reduction in CRPC

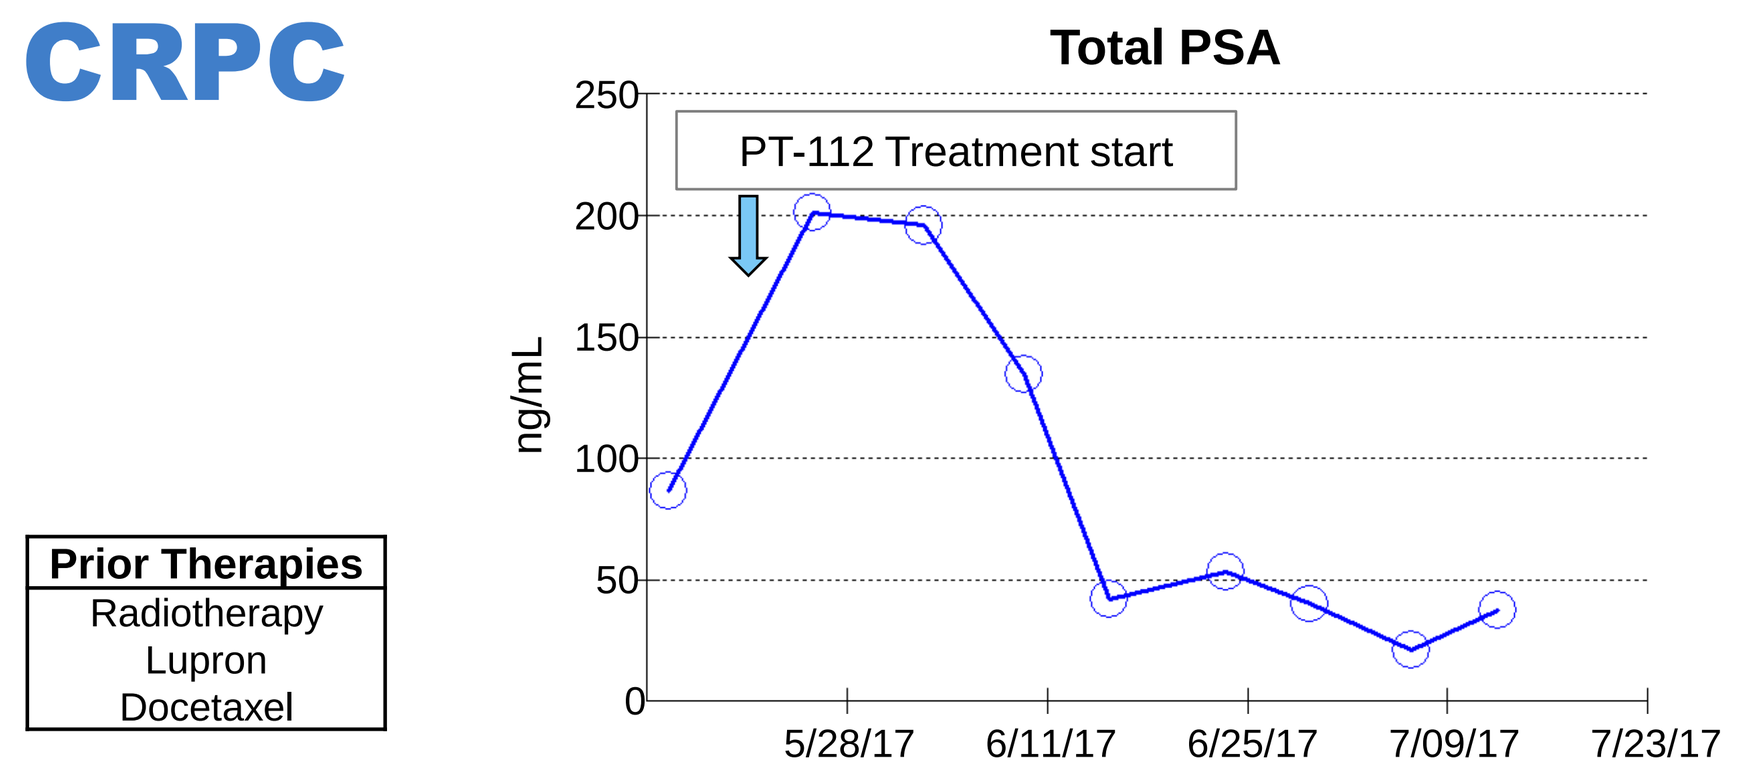

Total PSA
PT-112 Treatment start
Prior Therapies
Radiotherapy
Lupron
Docetaxel
ng/mL
0
50
100
150
200
250
5/28/17
6/11/17
6/25/17
7/09/17
7/23/17

Figure 6: PSA Response with PT-112 Treatment. A castration-resistant prostate cancer (CRPC) patient dosed at 200 mg/m^2 had a 78% decrease in serum PSA while on PT-112 treatment.

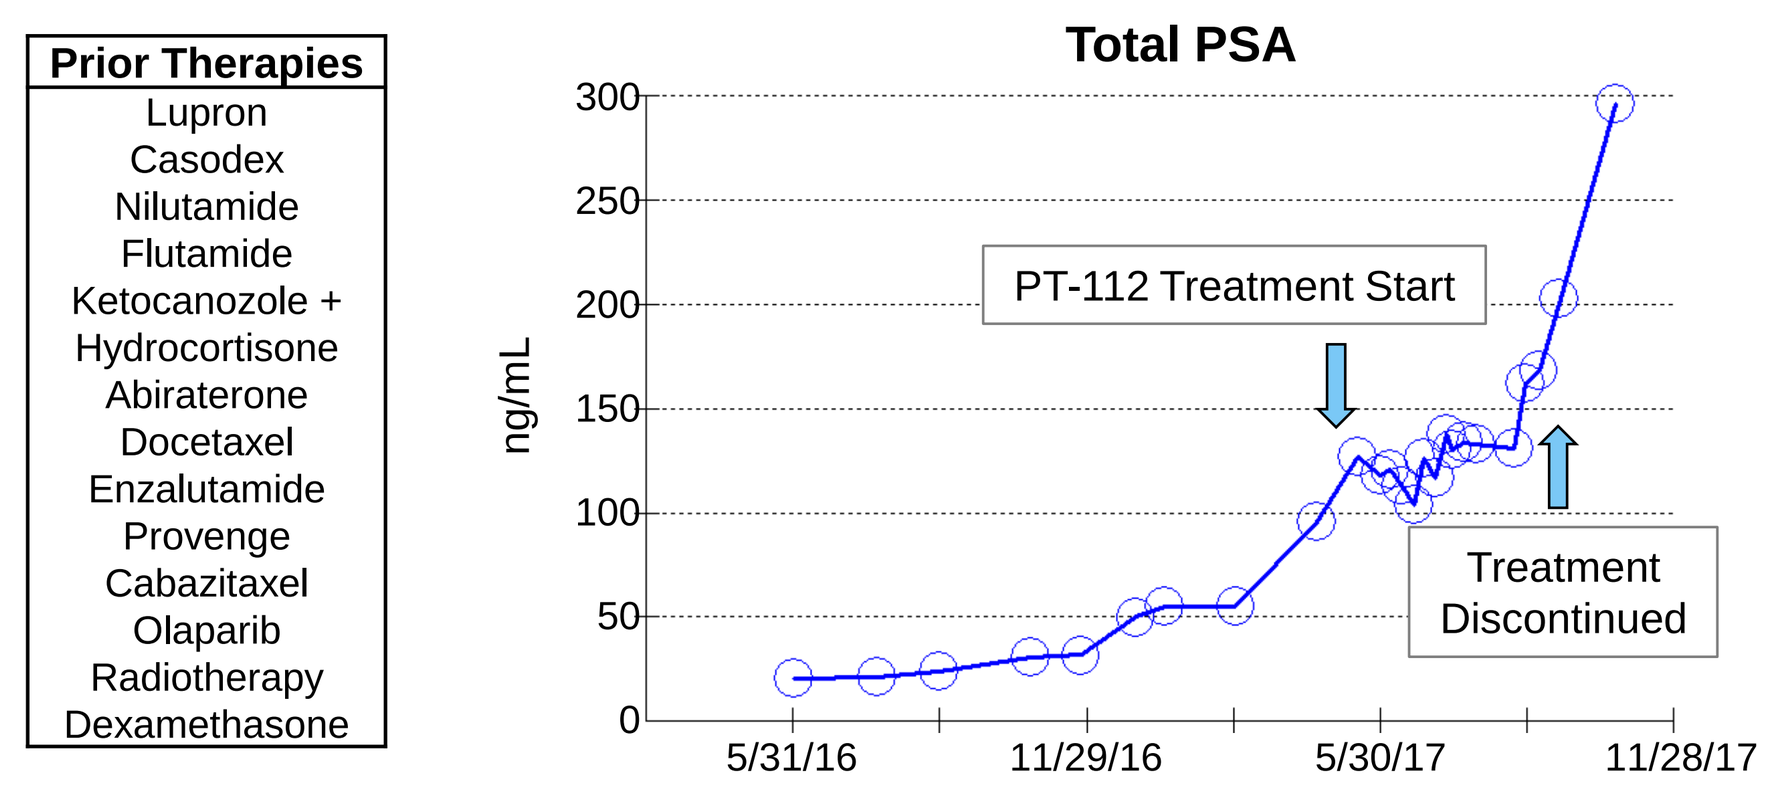

Prior Therapies
Lupron
Casodex
Nilutamide
Flutamide
Ketocanozole +
Hydrocortisone
Abiraterone
Docetaxel
Enzalutamide
Provenge
Cabazitaxel
Olaparib
Radiotherapy
Dexamethasone
Total PSA
PT-112 Treatment Start
Treatment Discontinued
ng/mL
0
50
100
150
200
250
300
5/31/16
11/29/16
5/30/17
11/28/17

Figure 7: PSA Control with PT-112 Treatment. A CRPC patient dosed at 250 mg/m^2 had a pronounced decrease in PSA velocity (and initial 20% reduction), corresponding with his time on PT-112 treatment. Additional patients experienced serum biomarker reductions (data not shown).

Pharmacokinetics

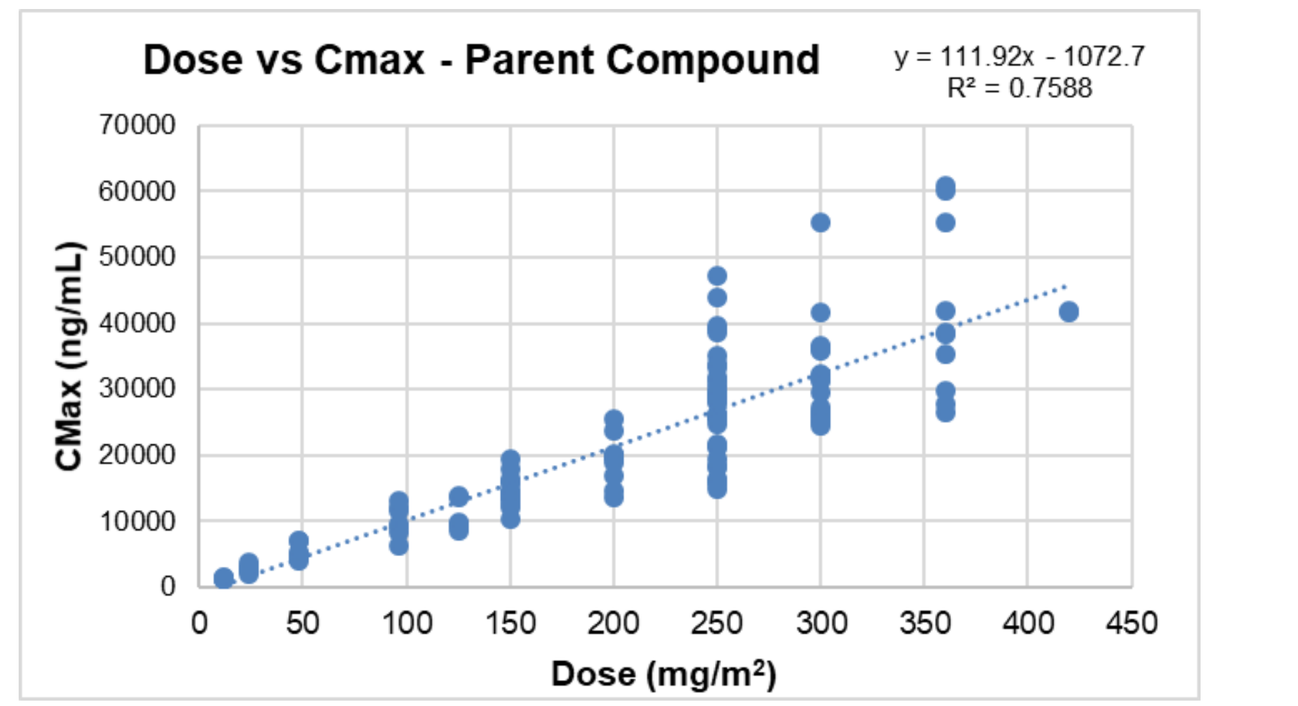

Dose vs Cmax - Parent Compound
y = 111.92x - 1072.7
R² = 0.7588
CMax (ng/mL)
0
10000
20000
30000
40000
50000
60000
70000
0
50
100
150
200
250
300
350
400
450
Dose (mg/m²)

Figure 8: Pharmacokinetics of PT-112. Parent compound in plasma was measured using 13 samples on days 1 and 8 of cycle 1, and 3 samples on day 1 of cycle 2. C_{max} and AUC_{inf} (data not shown) increase proportionally with escalating doses. Elemental Pt in plasma and plasma ultrafiltrate exhibits similar dose-proportional trends (data not shown).

Conclusions

- **PT-112 is well tolerated with clear signs of anti-cancer activity in heavily pre-treated patients crossing numerous solid tumor types**
- **Activity across a wide range of dose levels indicates a broad, positive therapeutic index**
- **Durable PRs / long-lasting PFS after treatment completion, with no additional therapy, were observed with PT-112 monotherapy in non-responders to prior immune checkpoint inhibitors**
- **Dramatic and durable PR in thymoma is indicative of potential in this rare disease**
- **In conjunction with preclinical data demonstrating potential best-in-class induction of ICD, these findings are consistent with PT-112 promoting an adaptive immune response in patients**
- **Further development in CRPC is also warranted; reductions in PSA or other biomarkers in 5/10 heavily pre-treated CRPC pts may be partly related to biodistribution to bone tissues**
- **PT-112's safety and efficacy profile make it a rational candidate for continued development, both as single agent and in combination therapy**
- **Based on clinical responses, as well as ICD induction and synergy with CPIs in nonclinical models, the combination of PT-112 with the anti-PD-L1 antibody avelumab is being studied in the Phase I / II PAVE-1 study (see NCT 03409458)**

References

1. Ames, T. D. *et al.* Findings across pre-clinical models in the development of PT-112, a novel investigational platinum-pyrophosphate anti-cancer agent. *European Journal of Cancer* 69, S153 (2016).
2. Ames, T. D. *et al.* Translational Research of PT-112, a Clinical Agent in Advanced Phase I Development: Evident Bone Tropism, Synergy In Vitro with Bortezomib and Lenalidomide , and Potent Efficacy in the Vk*MYC Mouse Model of Multiple Myeloma. *Blood* 130, 1797–1797 (2017).
3. Yamazaki, T. *et al.* Potent induction of immunogenic cell death by PT-112. Poster presentation at CRI-CIMT-EATI-AACR International Cancer Immunotherapy Conference (2018).
4. Karp, D. D. *et al.* A phase I study of PT-112 in advanced solid tumors. *JCO* 35, 2519–2519 (2017).
5. Tsao, A. S. et al. A practical guide of the Southwest Oncology Group (SWOG) to measure malignant pleural mesothelioma (MPM) tumors by RECIST and modified RECIST criteria. *J Thorac Oncol* 6, 598–601 (2011).

Contact authors at tames@phosplatin.com